IN VITRO DRUG TRANSPORTER INTERACTION STUDIES WITH THE OUTER MEMBRANE PROTEIN TARGETING ANTIBIOTIC MUREPAVADIN (POL7080)

G.J. Douglas[1], A. Klukovits[2], E. Nagy[2], Z. Gyömbör[2], L. Beni[1], C. Zwingelstein[1], G.E. Dale[1], **A. Wach**[1]
[1]Polyphor, 4123 Allschwil, Switzerland; [2]Solvo Biotechnology, Central Hungarian Innovations Center, Gyár utca 2, 2040 Budaörs, Hungary

POLYPHOR

Background

Murepavadin represents the first member of a novel class of outer membrane protein targeting antibiotics, being developed by Polpyhor for the treatment of serious infections caused by *Pseudomonas aeruginosa* (*P. aeruginosa*). Murepavadin binds to and inhibits an outer membrane protein of intact bacteria, lipopolysaccharide transport protein D (LptD), which is involved in lipopolysaccharide biogenesis in Gram-negative bacteria.

Murepavadin is mainly active against *P. aeruginosa* and no significant effect is seen on other tested species with the exception of *P. stutzeri* and *P. mendocina*. Thus, unlike broad-spectrum antibiotics, treatment with murepavadin would not generate resistance or negatively impact the native flora of the subject. Murepavadin has demonstrated potent antibacterial activity in vitro (MIC_{90} was 0.125 µg/mL when tested on >1200 *P. aeruginosa* isolates from the United States, Europe, and China) and in mouse models of septicemia, peritonitis, and lung and thigh infections.

Objective

To investigate the potential for drug-drug interactions induced by inhibition of drug transporter mechanisms.

The effect of murepavadin on probe substrate uptake by a number of hepatic and renal transporters known to be involved in clinically relevant drug interactions was investigated.

Methods

- Cellular uptake of radiolabeled probe substrates was measured in CHO, MDCK or HEK293 cells stably expressing (OAT2 transient expression) drug transporters in the presence or absence of murepavadin (0.4-300 µM) or reference transporter inhibitor
- Uptake of probe substrate in control cells not expressing transporters provided background activity values.

Results

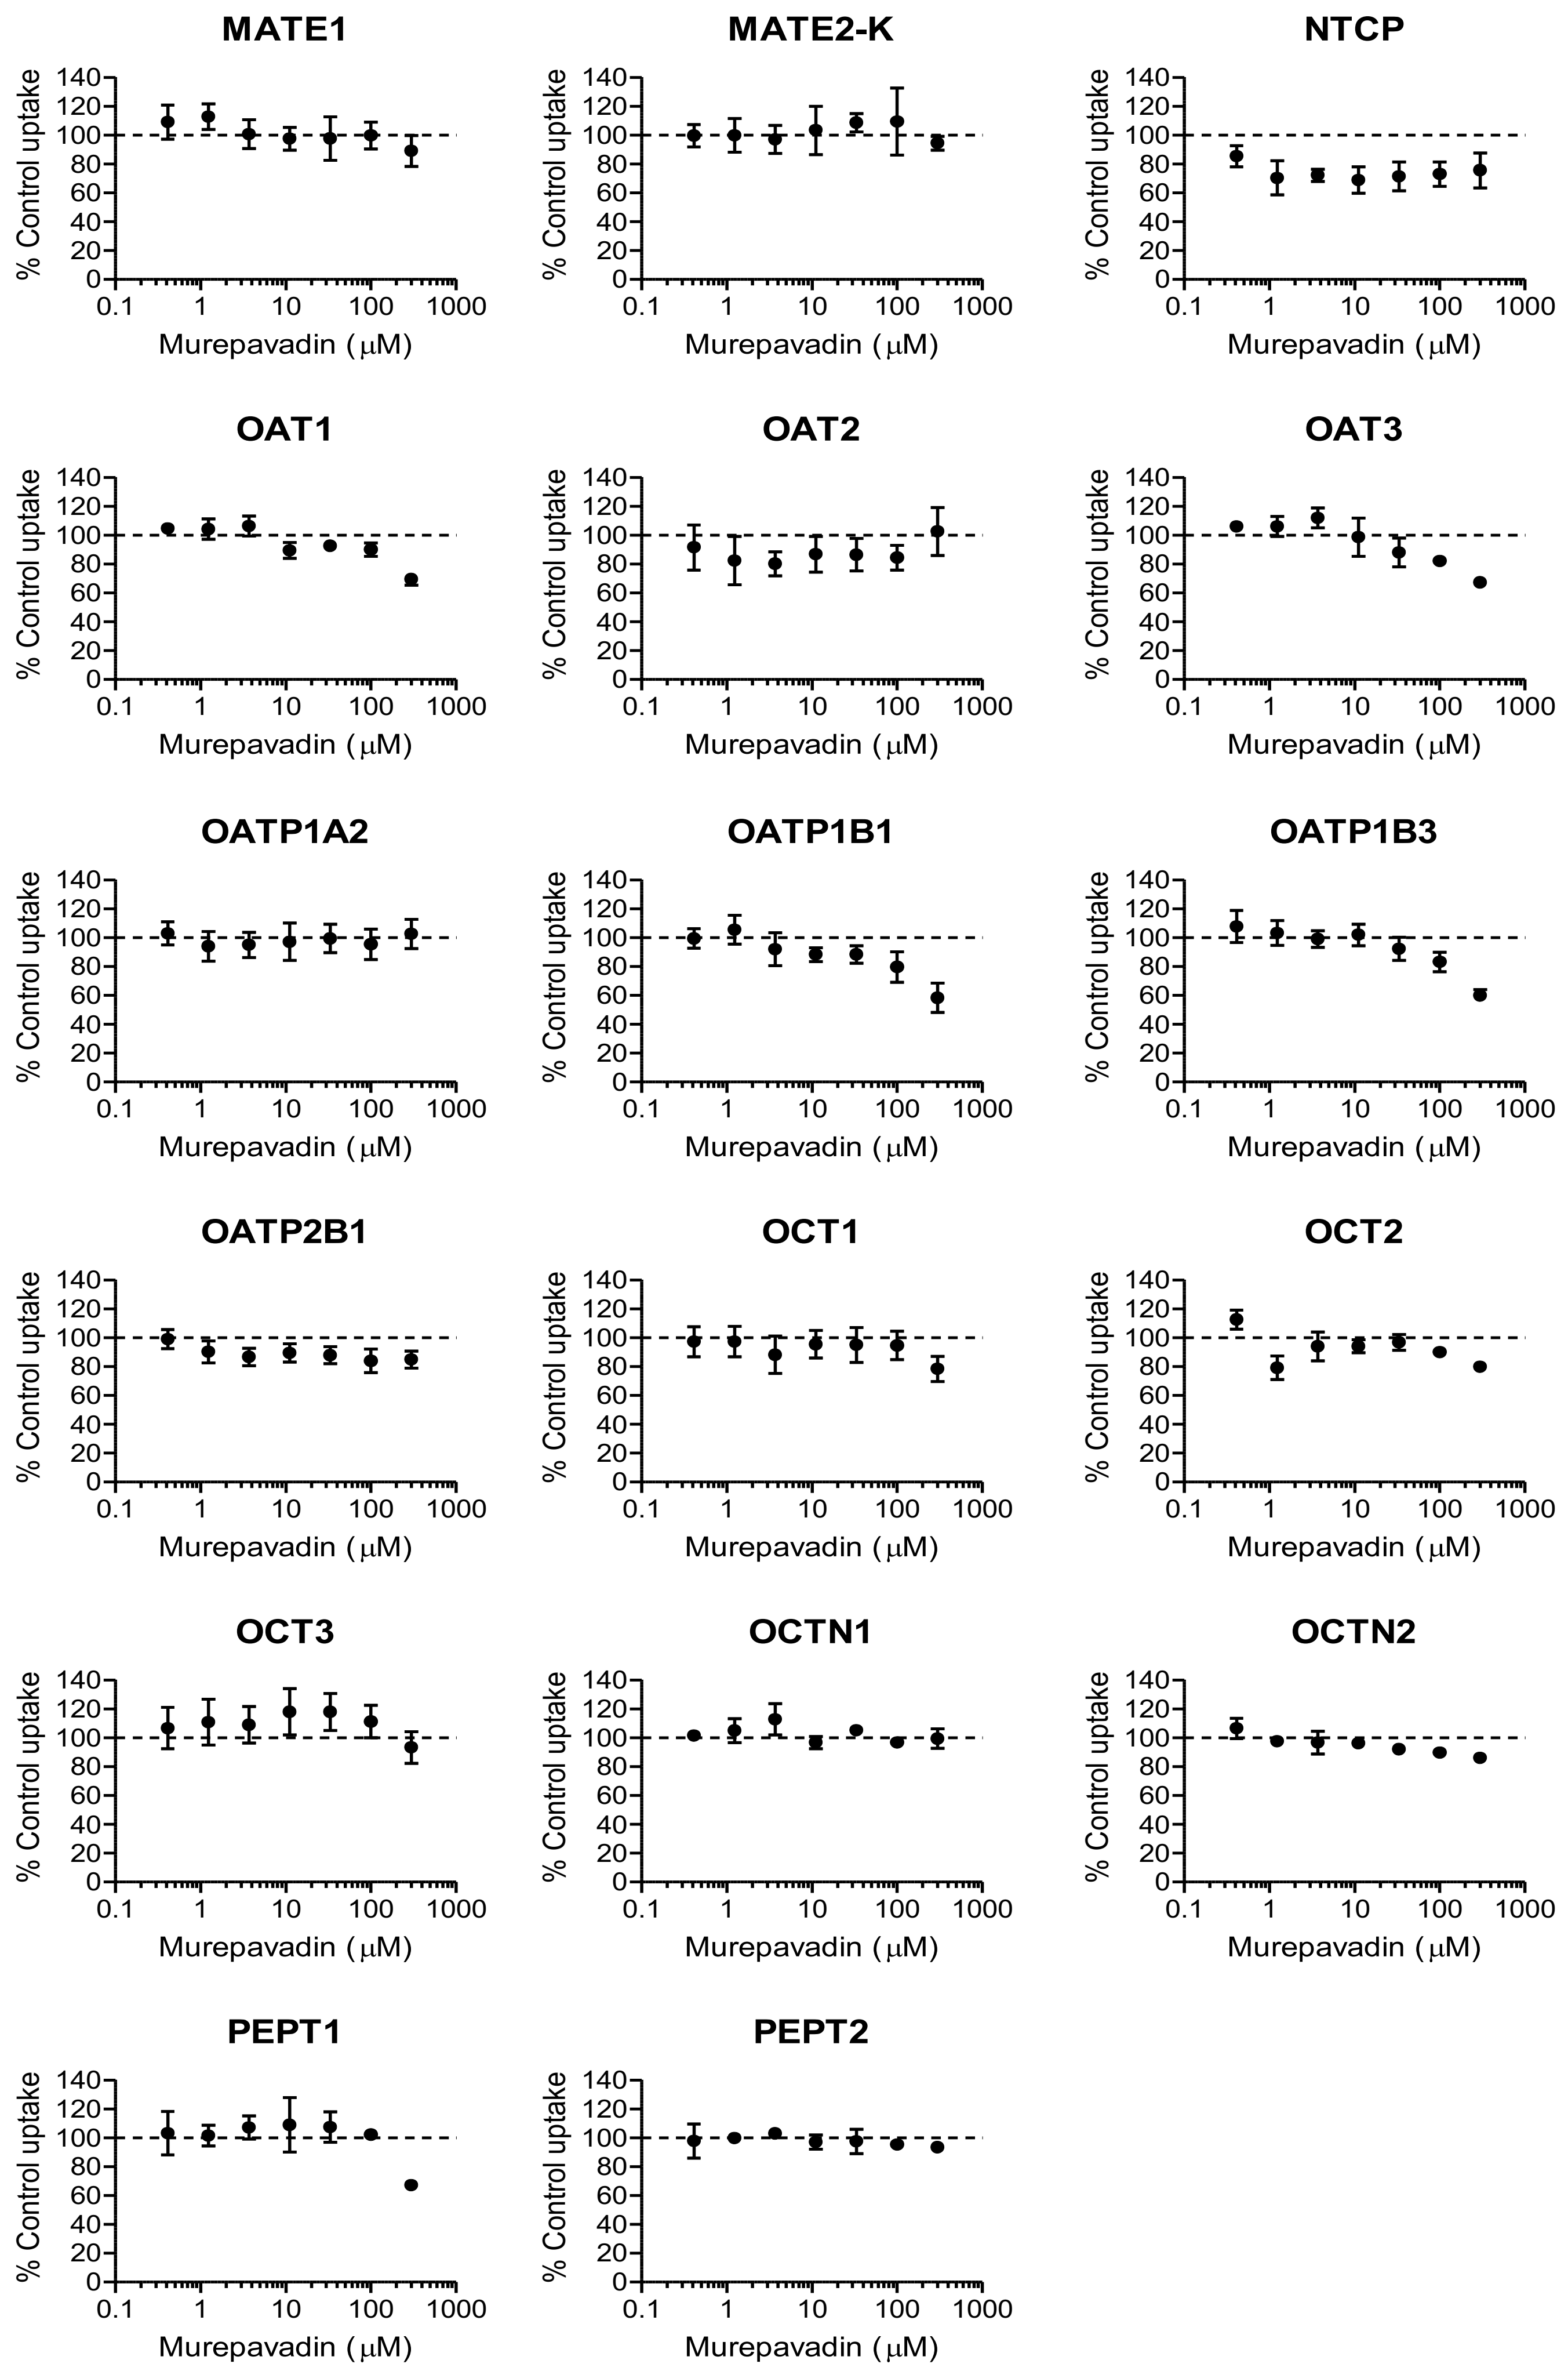
MATE1
MATE2-K
NTCP
OAT1
OAT2
OAT3
OATP1A2
OATP1B1
OATP1B3
OATP2B1
OCT1
OCT2
OCT3
OCTN1
OCTN2
PEPT1
PEPT2
% Control uptake
Murepavadin (µM)

- Reference drugs inhibited probe substrate uptake by 74-100%.
- Murepavadin did not significantly inhibit probe substrate accumulation by MATE-1, MDR1, OAT2, OATP1A2, OATP2B1, OCT2, OCT3, OCTN1, OCTN2 or PEPT2.
- At 300 µM murepavadin inhibited NTCP, OAT1, OAT3, OATP1B1, OATP1B3 and OCT1 and PEPT1 to some extent, but it was not possible to calculate IC50 values.
- In addition, murepavadin did not inhibit digoxin permability or efflux by Caco-2 monolayers.

Table 1: XYZ

Transporter	Main function	Probe substrate	Reference inhibitor	300 µM murepavidin (% inhibition)
NTCP	Hepatic uptake	Taurocholate 2 µM	TCDC 100 µM	24%
OAT1	Renal uptake	PAH 1.33 µM	Benzbromarone 200 µM	30%
OAT3	Renal uptake	E3S 1.0 µM	Probenecid 200 µM	33%
OATP1B1	Hepatic uptake	E3S 0.1 µM	Cerivastatin 100 µM	42%
OATP1B3	Hepatic uptake	Fluo-3 10 µM	Fluvastatin 30 µM	40%
OATP2B1	Hepatic uptake	E3S 0.2 µM	Fluvastatin 10 µM	16%
OCT1	Hepatic uptake	Metformin 10 µM	Verapamil 100 µM	22%
PEPT1	Renal reuptake	Gly-Sar 1.8 µM	tyr-Phe 200 µM	30%

Conclusion

- In the intended patient population the steady state Cmax of murepavadin was between 3.0-6.6 µM at therapeutic doses, substantially below the concentrations where any inhibition was observed.
- Thus, at predicted clinical exposures murepavadin is unlikely to cause drug-drug interactions with substrates of the transporters studied.

ECCMID 2017 Vienna, Poster 2822